AACR Poster #1715: Anti-cancer activity of a SN-38 nanoparticle, DEP® irinotecan, in human colon and pancreatic cancer xenograft models

David J Owen[1], Brian D. Kelly[1], Victoria McLeod[2], Rachael Walker[2], Jeannette Schreuders[2], Susan Jackson[2], Michael Giannis[1], Christine Dietinger[1], Shirley Xia[1], Anne Cargill[1], Aynaz Seta[1], Richard Hufton[1], Graham Heery[1], Carleen Cullinane[2] 1. Starpharma Pty Ltd, Melbourne, Australia, 2. Peter MacCallum Cancer Centre, Melbourne, Australia

Contact:
David.owen@starpharma.com or www.starpharma.com

INTRODUCTION

Starpharma's novel polylysine dendrimer-based DEP® platform has broad commercial applicability in drug delivery by enhancing the therapeutic utility of drugs through improved solubility, efficacy and pharmacokinetics, reductions in certain toxicities (e.g. bone marrow toxicity) and generation of new intellectual property. The novel DEP platform has shown reproducible advantages across a wide range of drug classes and can be utilised with both small molecule drugs, peptides and proteins. Currently there are four DEP candidates in the clinic; DEP docetaxel, DEP cabazitaxel, and DEP irinotecan. The fourth DEP candidate, AZD0466, is a promising Bcl-2/Bcl-xL inhibitor, partnered with AstraZeneca, and is in Phase 1 clinical trials in the US (See AACR Poster # P-56, P-1718 & Abs 3066).

Conventional irinotecan (Campostar®) is a pro-drug, which, following intravenous administration, needs to be converted in the liver to the active anti-cancer agent, known as SN-38 (**Figure 1**). In contrast, Starpharma's DEP irinotecan (Starpharma's third internal DEP® candidate to enter the clinic) incorporates the active irinotecan derivative SN-38, avoiding the need for hepatic conversion and avoiding the variability in SN-38 for therapeutic effect. DEP irinotecan is expected to accumulate preferentially in tumour tissue to exert its superior anti-tumour effect, as is seen with DEP docetaxel, DEP cabazitaxel and other DEP conjugates.

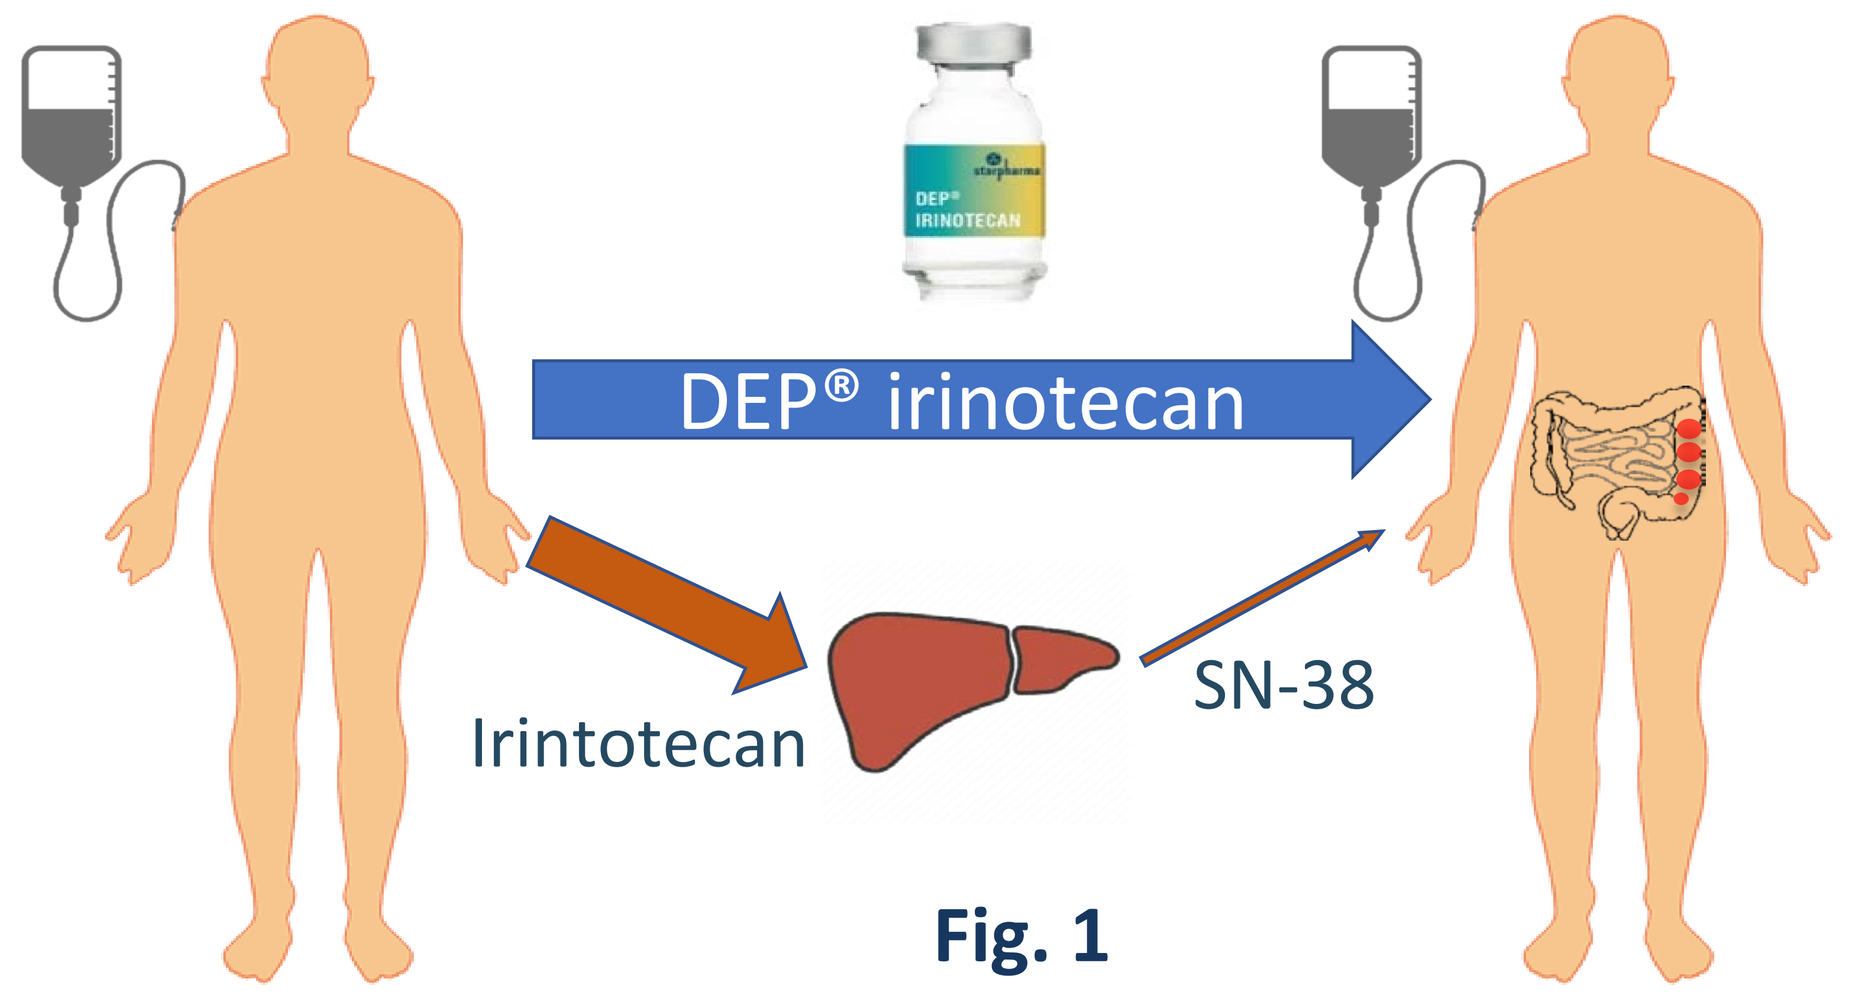
DEP® irinotecan
Irintotecan
SN-38

Fig. 1

METHODS

Balb/c nude mice were inoculated subcutaneously with a cancer cell line

- Exp 1 – HT-29 or SW620; 10 and 6 mice/group respectively
- Exp 2 –HT-29; 8 mice/group
- Exp 3 –HT-29; 10 mice/group

Mice were dosed with saline as control and;

- Exp 1 – DEP irinotecan (15mg/kg) and irinotecan (90mg/kg) IV once per week on days 1, 8 and 15 (all drug groups were dosed at the pre-determined maximum tolerated dose for each therapy).
- Exp 2 – DEP irinotecan (low and high dose), and irinotecan (35 mg/kg) IV once per week and cetuximab (Erbitux® 25 mg/kg) IP twice per week. Irinotecan and cetuximab (Erbitux®) were dosed at the pre-determined maximum tolerated dose for the combination; however, DEP irinotecan doses were low dose (5 mg/kg) and high dose (10 mg/kg) of the maximum tolerated dose for this combination.
- Exp 3 – DEP irinotecan (low and high dose), and irinotecan (80 mg/kg) IV once per week. Olaparib (50 mg/kg, Lynparza®) dosed PO (per oral) five times per week (5 days on/2 days off). Irinotecan and olaparib were dosed at the pre-determined maximum tolerated dose for the combination; however, DEP irinotecan doses were deliberately reduced in this experiment to allow for demonstration of synergy and were low dose (5 mg/kg) and high dose (8 mg/kg) of the maximum tolerated dose of single agent when used in this combination.

Tumour growth data (mean ± standard error of the mean (SEM)) were analysed in GraphPad Prism

- Exp 1 and Exp 2 – two-way ANOVA followed by Dunnett's post-hoc test. Kaplan-Meier survival curves were analysed using the Log-rank (Mantel-Cox) test.
- Exp 3 – two-way ANOVA with repeated measures followed by i) Dunnett's multiple comparison test to compare monotherapies to vehicle control and ii) Tukey's multiple comparisons test for comparisons between treatment groups.

(Note: If error bars do not display on the graphs, they are not visible because they are shorter than the height of the symbol).

DENDRIMER CONJUGATES AS THERAPEUTICS

Improved efficacy:

DEP® improves anti-cancer efficacy through better drug targeting & improved pharmacokinetics.

Benefits in combination:

DEP® drugs are ideal candidates for combination therapy including with immuno-oncology (IO) agents and other chemo. DEP® drugs show synergistic benefits over the original versions and given they have been shown to reduce bone marrow toxicity and do not require pre-treatment with cortisone, they are particularly well suited to combine with IO.

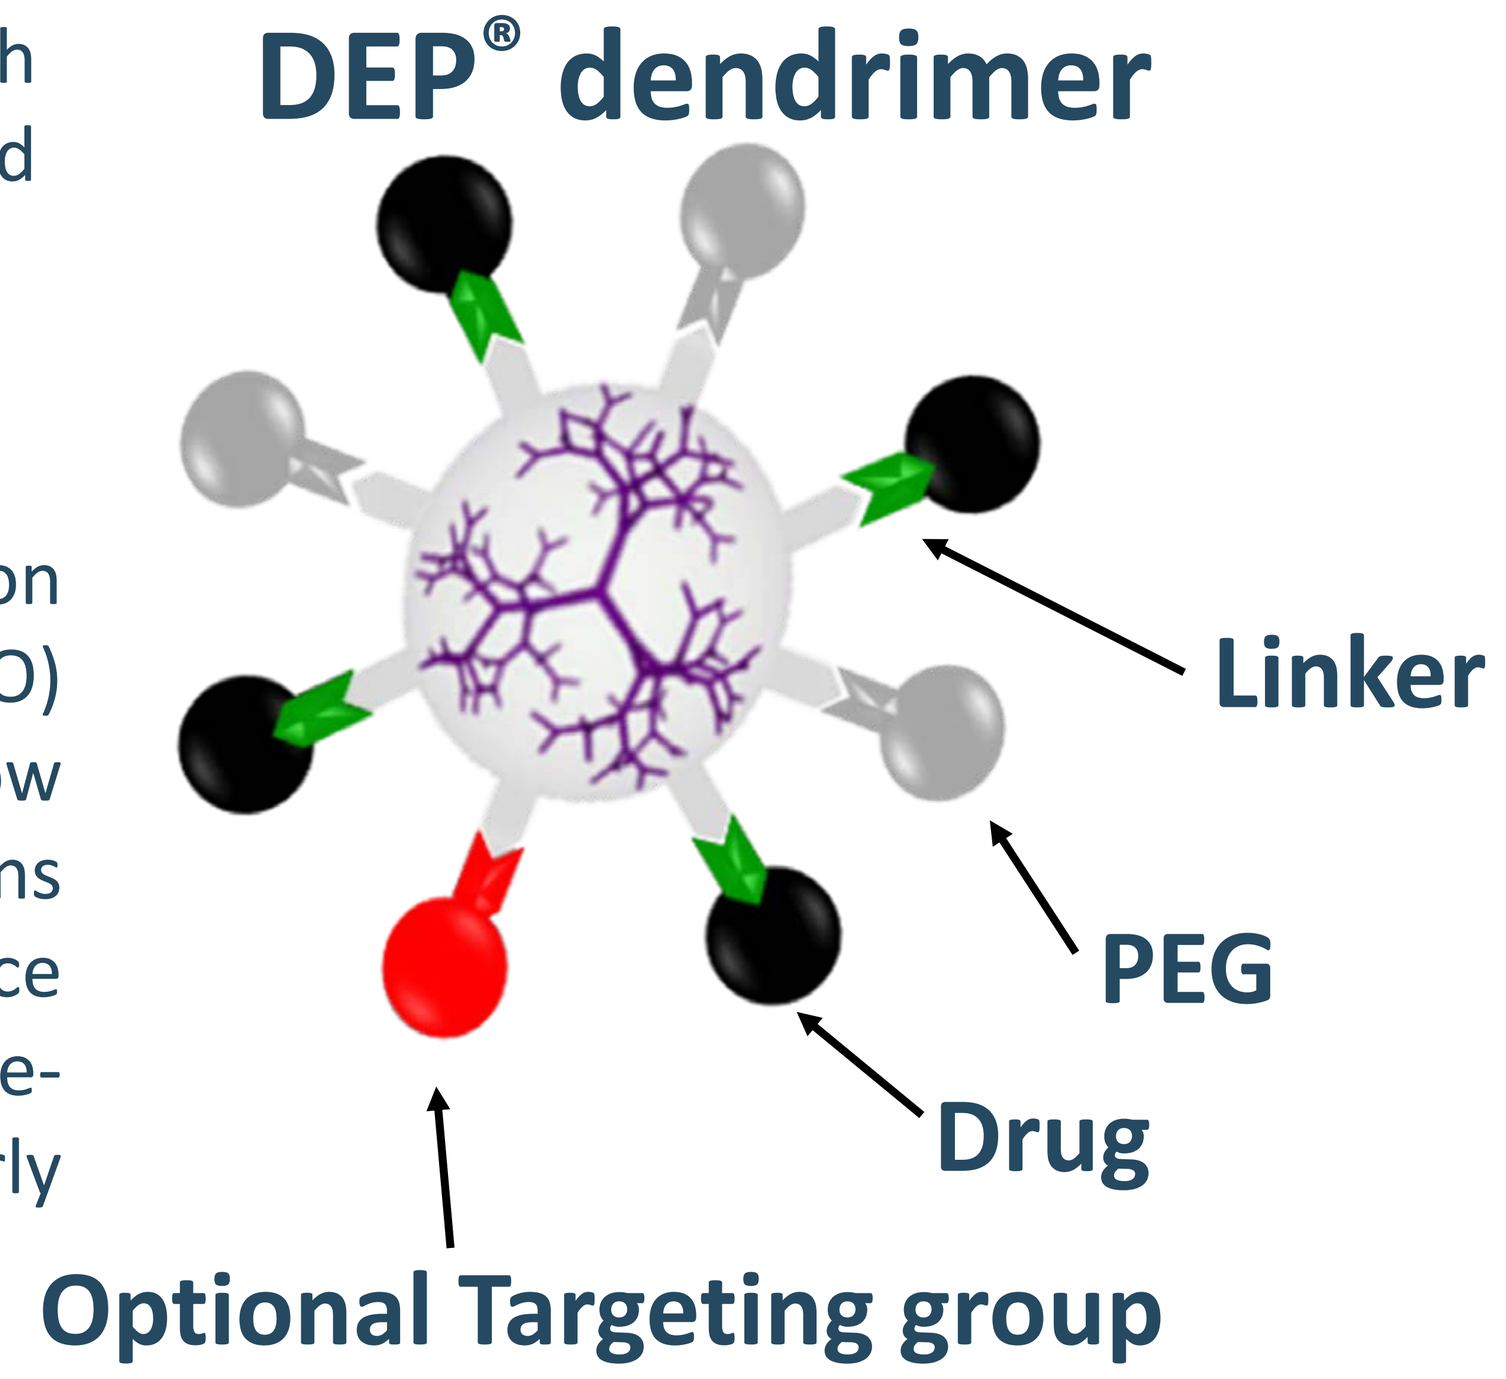
DEP® dendrimer
Linker
PEG
Drug
Optional Targeting group

Reduced side-effects:

DEP® reduces important side effects such as bone marrow toxicity / low white blood cells (neutropenia) and alopecia (hair loss). DEP® removes the need for toxic detergents in current formulations, which are highly soluble in aqueous formulations.

DEP IRINOTECAN MONOTHERAPY (EXPERIMENT 1)

Complete tumour regression and 100% survival was observed in DEP irinotecan treated animals bearing the colon cancer tumour cell line, SW620 (**Figures 2 and 3**). The tumour inhibition and survival effects of DEP irinotecan were markedly improved compared with the irinotecan treated group and the differences were highly statistically significant (P<0.0001 and P<0.0045, respectively)

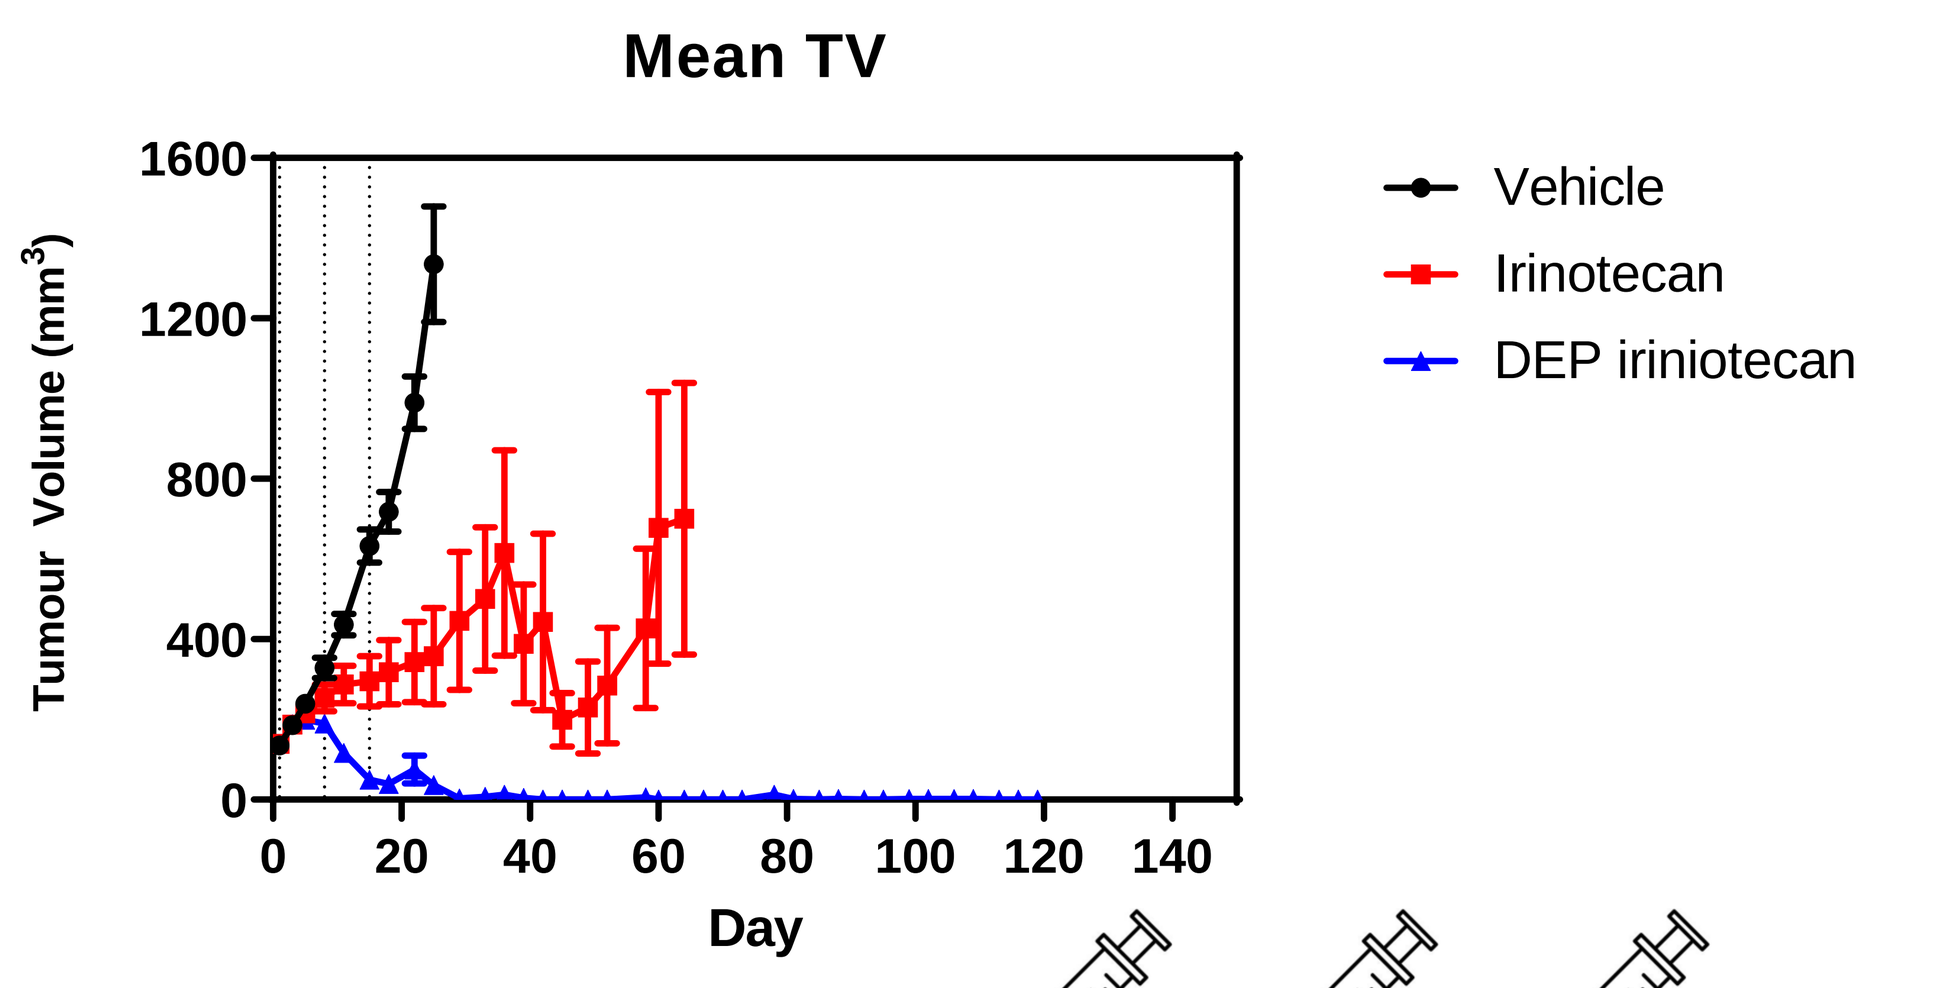
Mean TV
Tumour Volume (mm³)
Day
Vehicle
Irinotecan
DEP iriniotecan

Fig. 2

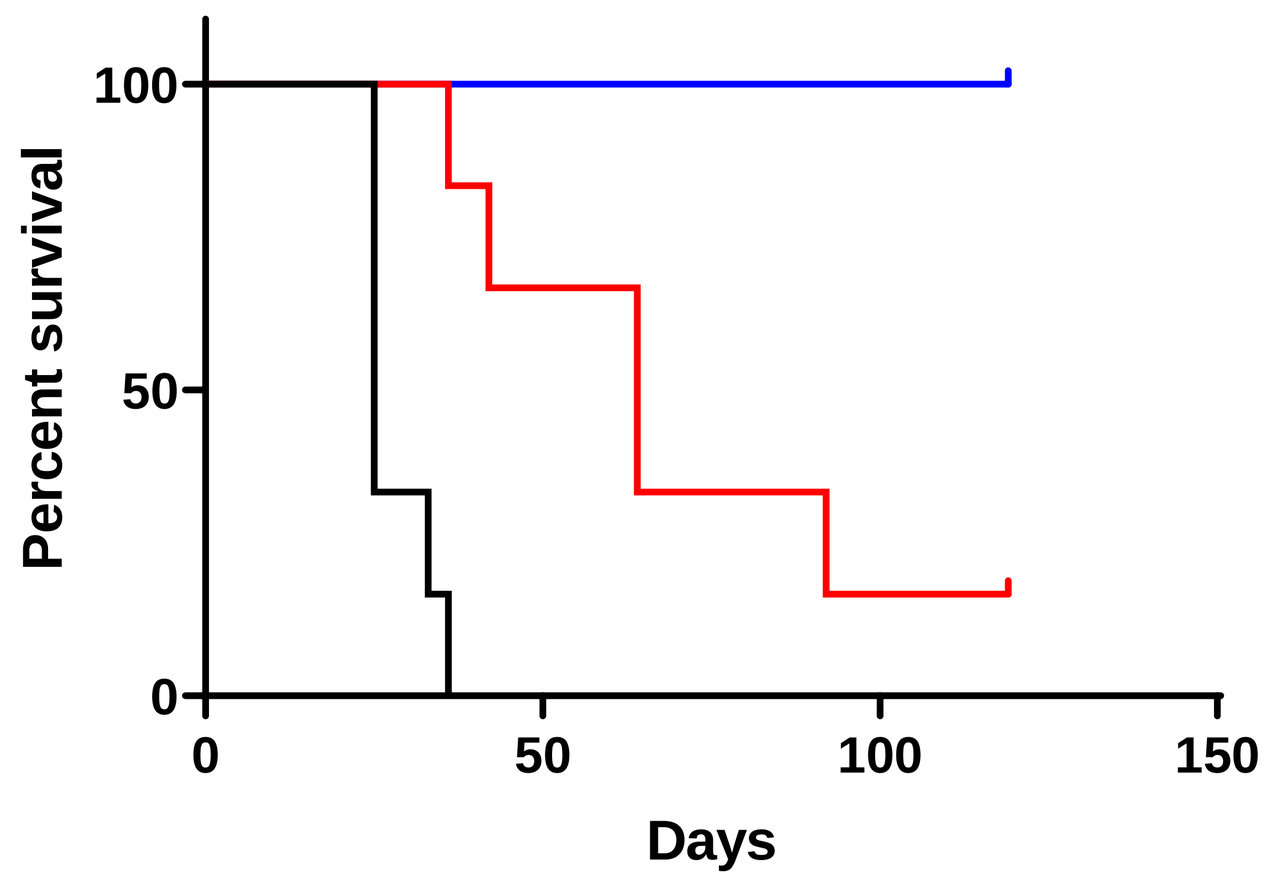
Percent survival
Days

Fig. 3

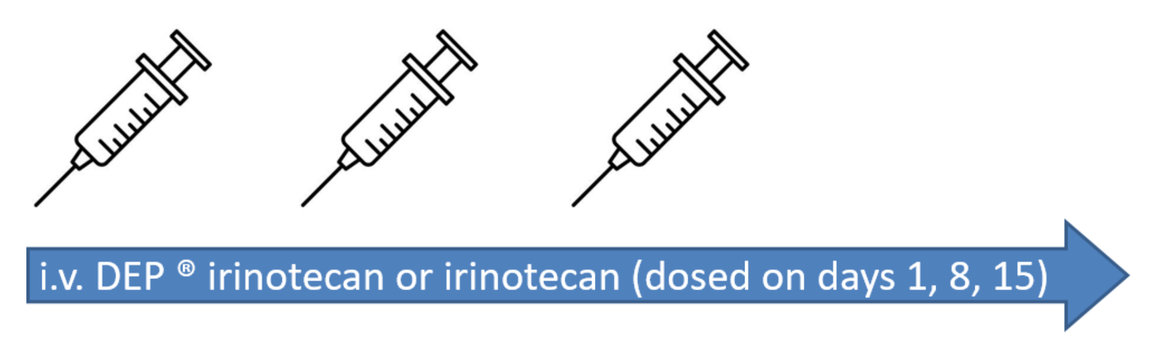
i.v. DEP ® irinotecan or irinotecan (dosed on days 1, 8, 15)

DEP irinotecan was also shown to be very effective in the HT-29 colon cancer model, which typically responds poorly to irinotecan.

In this study, irinotecan did not achieve appreciable anti-cancer activity compared to saline, whereas DEP irinotecan exhibited a significant anti-cancer effect (**Figure 4**). DEP irinotecan was significantly more effective than irinotecan (P<0.0001) for both enhanced efficacy and survival and was well tolerated in this model (see **Figures 4 and 5**).

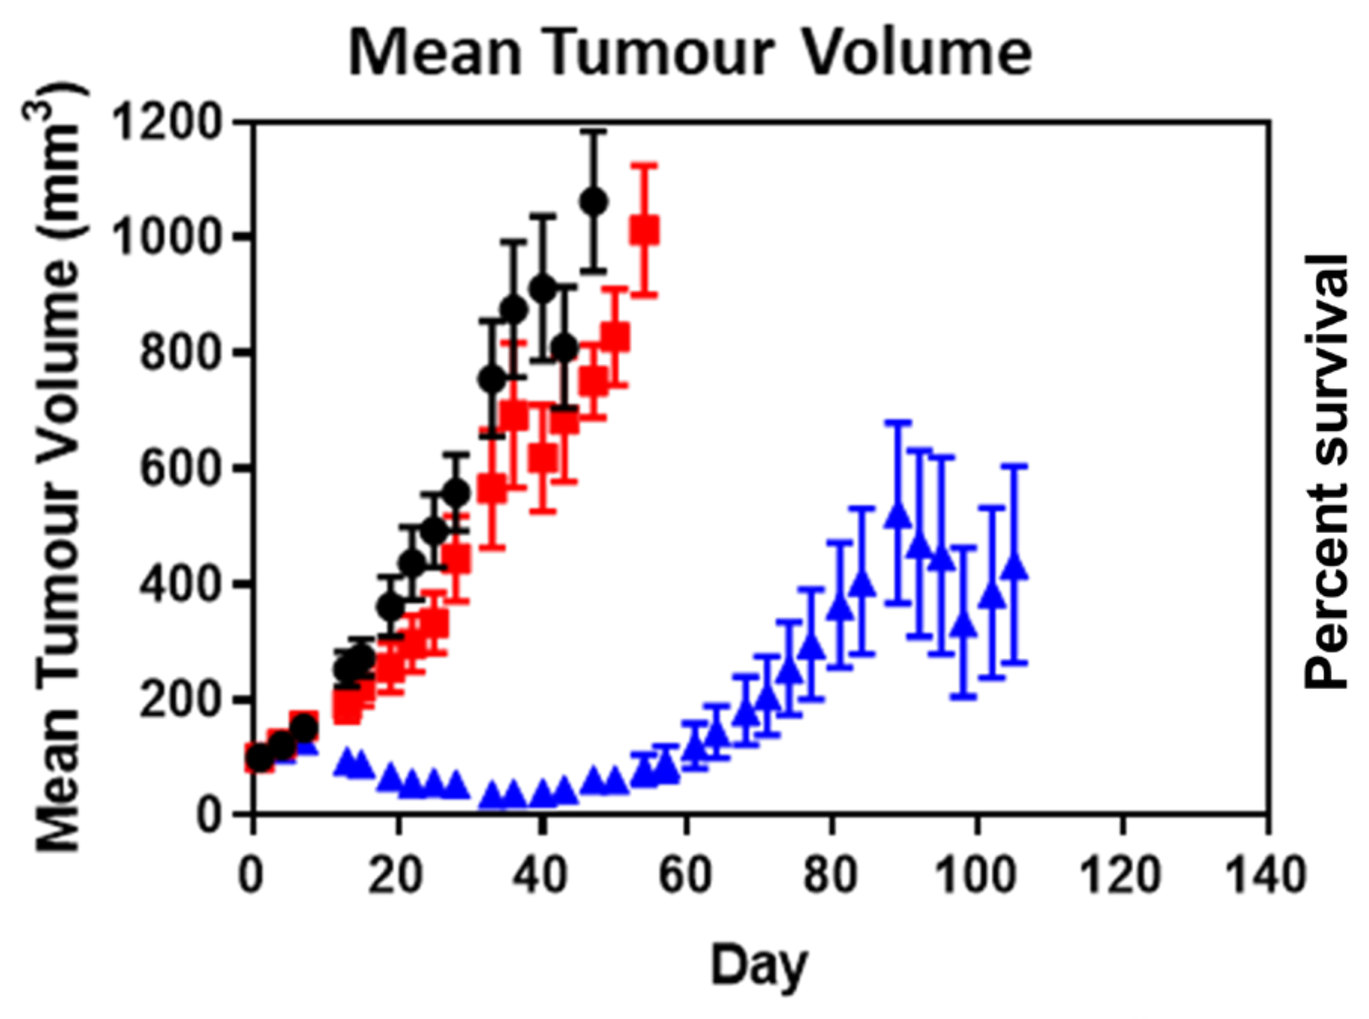
Mean Tumour Volume
Mean Tumour Volume (mm³)
Day

Fig. 4

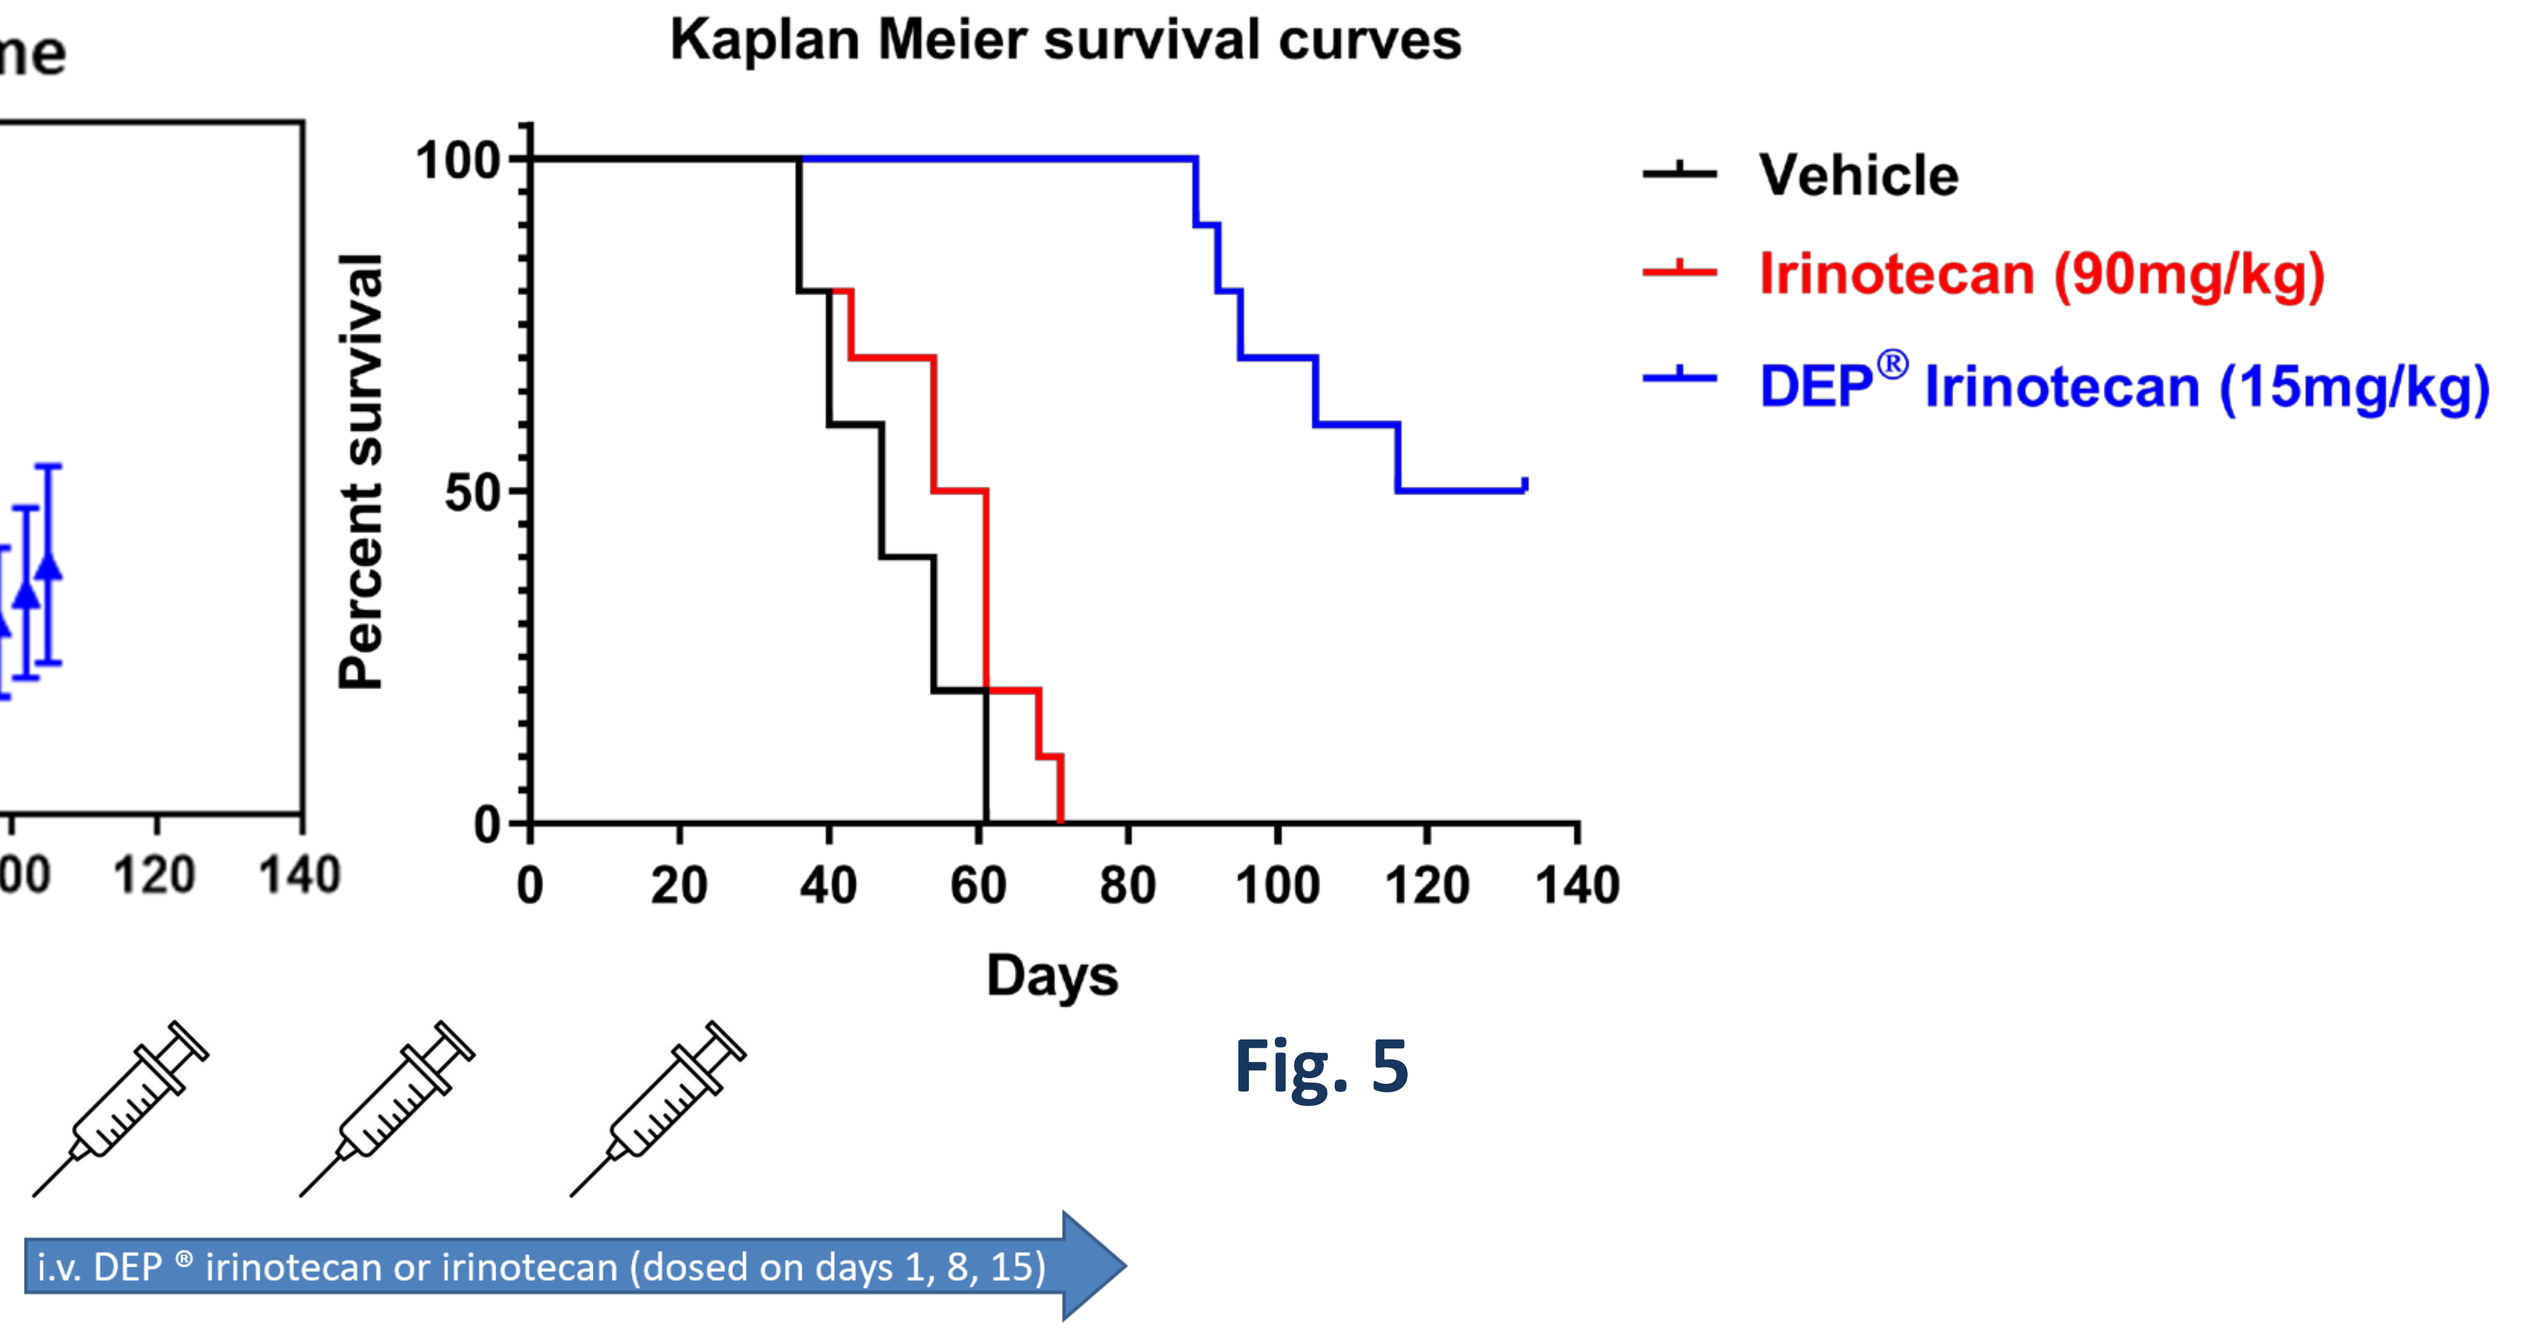
Kaplan Meier survival curves
Percent survival
Days
Vehicle
Irinotecan (90mg/kg)
DEP® Irinotecan (15mg/kg)
i.v. DEP ® irinotecan or irinotecan (dosed on days 1, 8, 15)

Fig. 5

DEP IRINOTECAN + CETUXIMAB (EXPERIMENT 2)

In the irinotecan-refractory human colon cancer model HT-29 xenograft, the combination of cetuximab (Erbitux®) and irinotecan displayed limited tumour inhibition (**Figures 6 and 7**). In contrast, DEP irinotecan in combination with cetuximab resulted in significantly enhanced anti-cancer efficacy and survival despite the DEP irinotecan doses being approximately one third (low dose) and approximately two thirds (high dose) of the maximum tolerated dose for this combination.

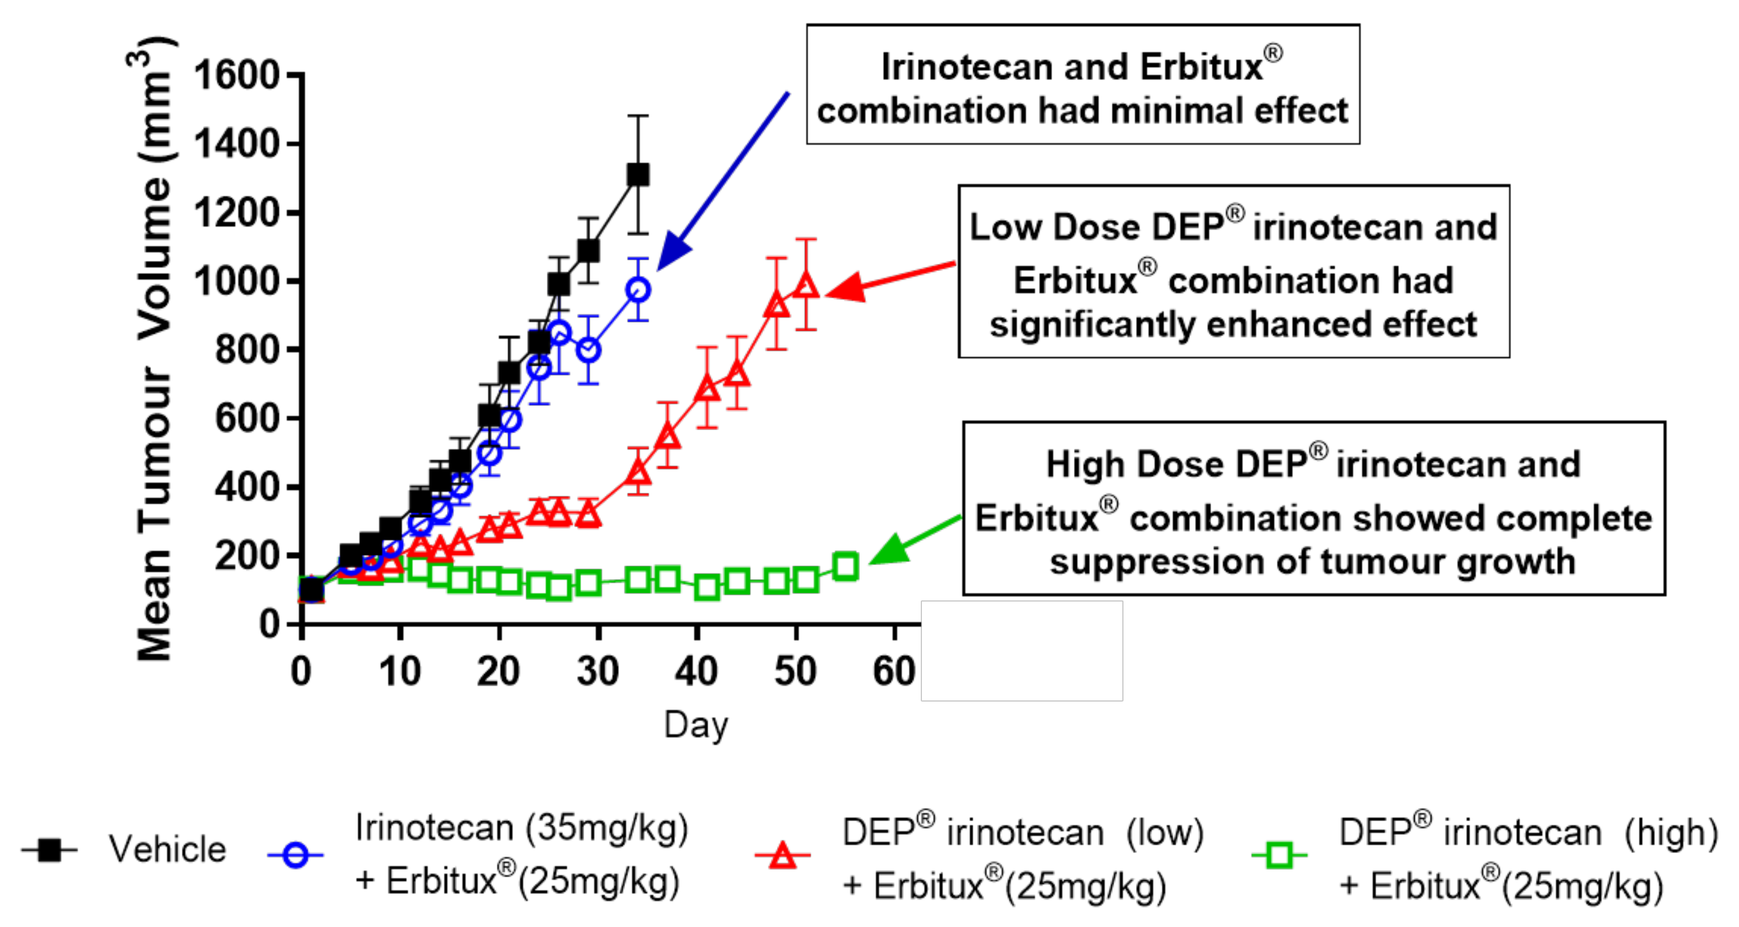
Mean Tumour Volume (mm³)
Day
Irinotecan and Erbitux® combination had minimal effect
Low Dose DEP® irinotecan and Erbitux® combination had significantly enhanced effect
High Dose DEP® irinotecan and Erbitux® combination showed complete suppression of tumour growth
Vehicle
Irinotecan (35mg/kg) + Erbitux®(25mg/kg)
DEP® irinotecan (low) + Erbitux®(25mg/kg)
DEP® irinotecan (high) + Erbitux®(25mg/kg)

Fig. 6

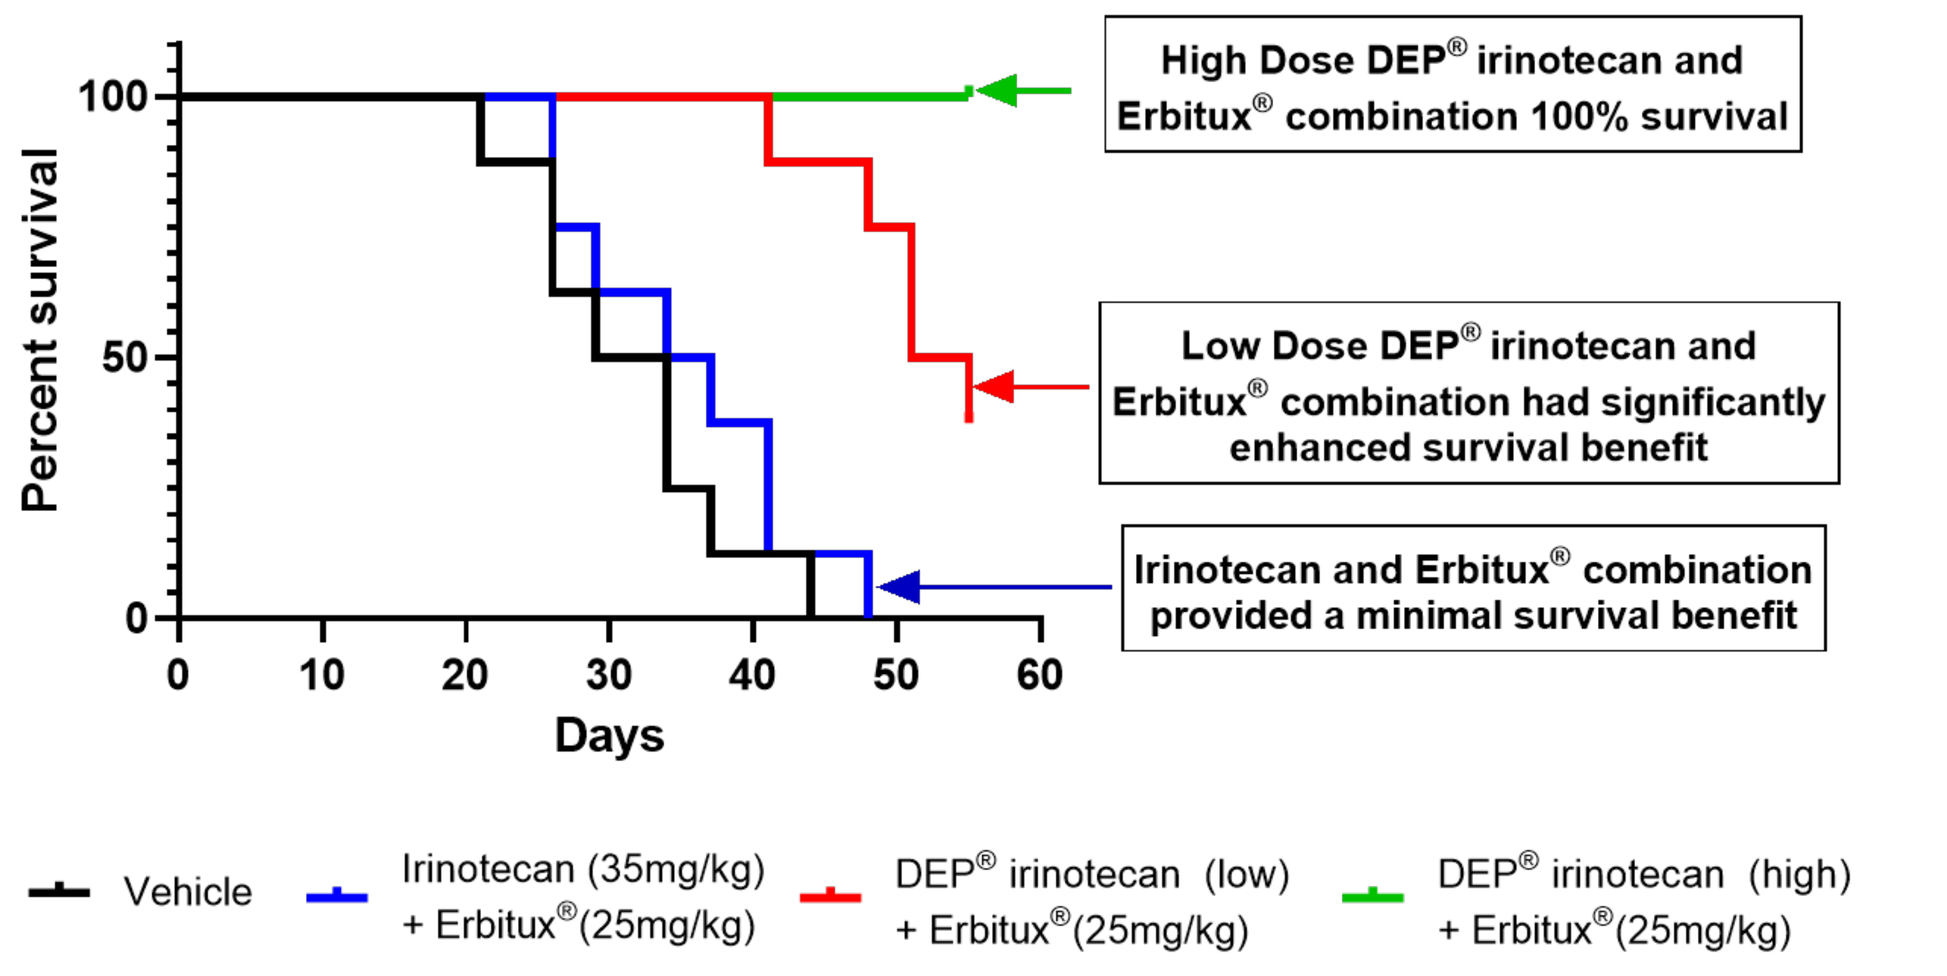
Percent survival
Days
High Dose DEP® irinotecan and Erbitux® combination 100% survival
Low Dose DEP® irinotecan and Erbitux® combination had significantly enhanced survival benefit
Irinotecan and Erbitux® combination provided a minimal survival benefit
Vehicle
Irinotecan (35mg/kg) + Erbitux®(25mg/kg)
DEP® irinotecan (low) + Erbitux®(25mg/kg)
DEP® irinotecan (high) + Erbitux®(25mg/kg)

Fig. 7

DEP IRINOTECAN + OLAPARIB (EXPERIMENT 3)

DEP irinotecan showed a significant and dose-related inhibition of growth in the irinotecan-refractory human colon cancer (HT-29) xenograft (**Figure 8**). At day 28, standard irinotecan showed modest tumour growth inhibition of 33% versus the vehicle control (P<0.05). Treatment with DEP irinotecan provided a significantly greater level of inhibition – 62% for low dose and 97% for high dose (P<0.0001).

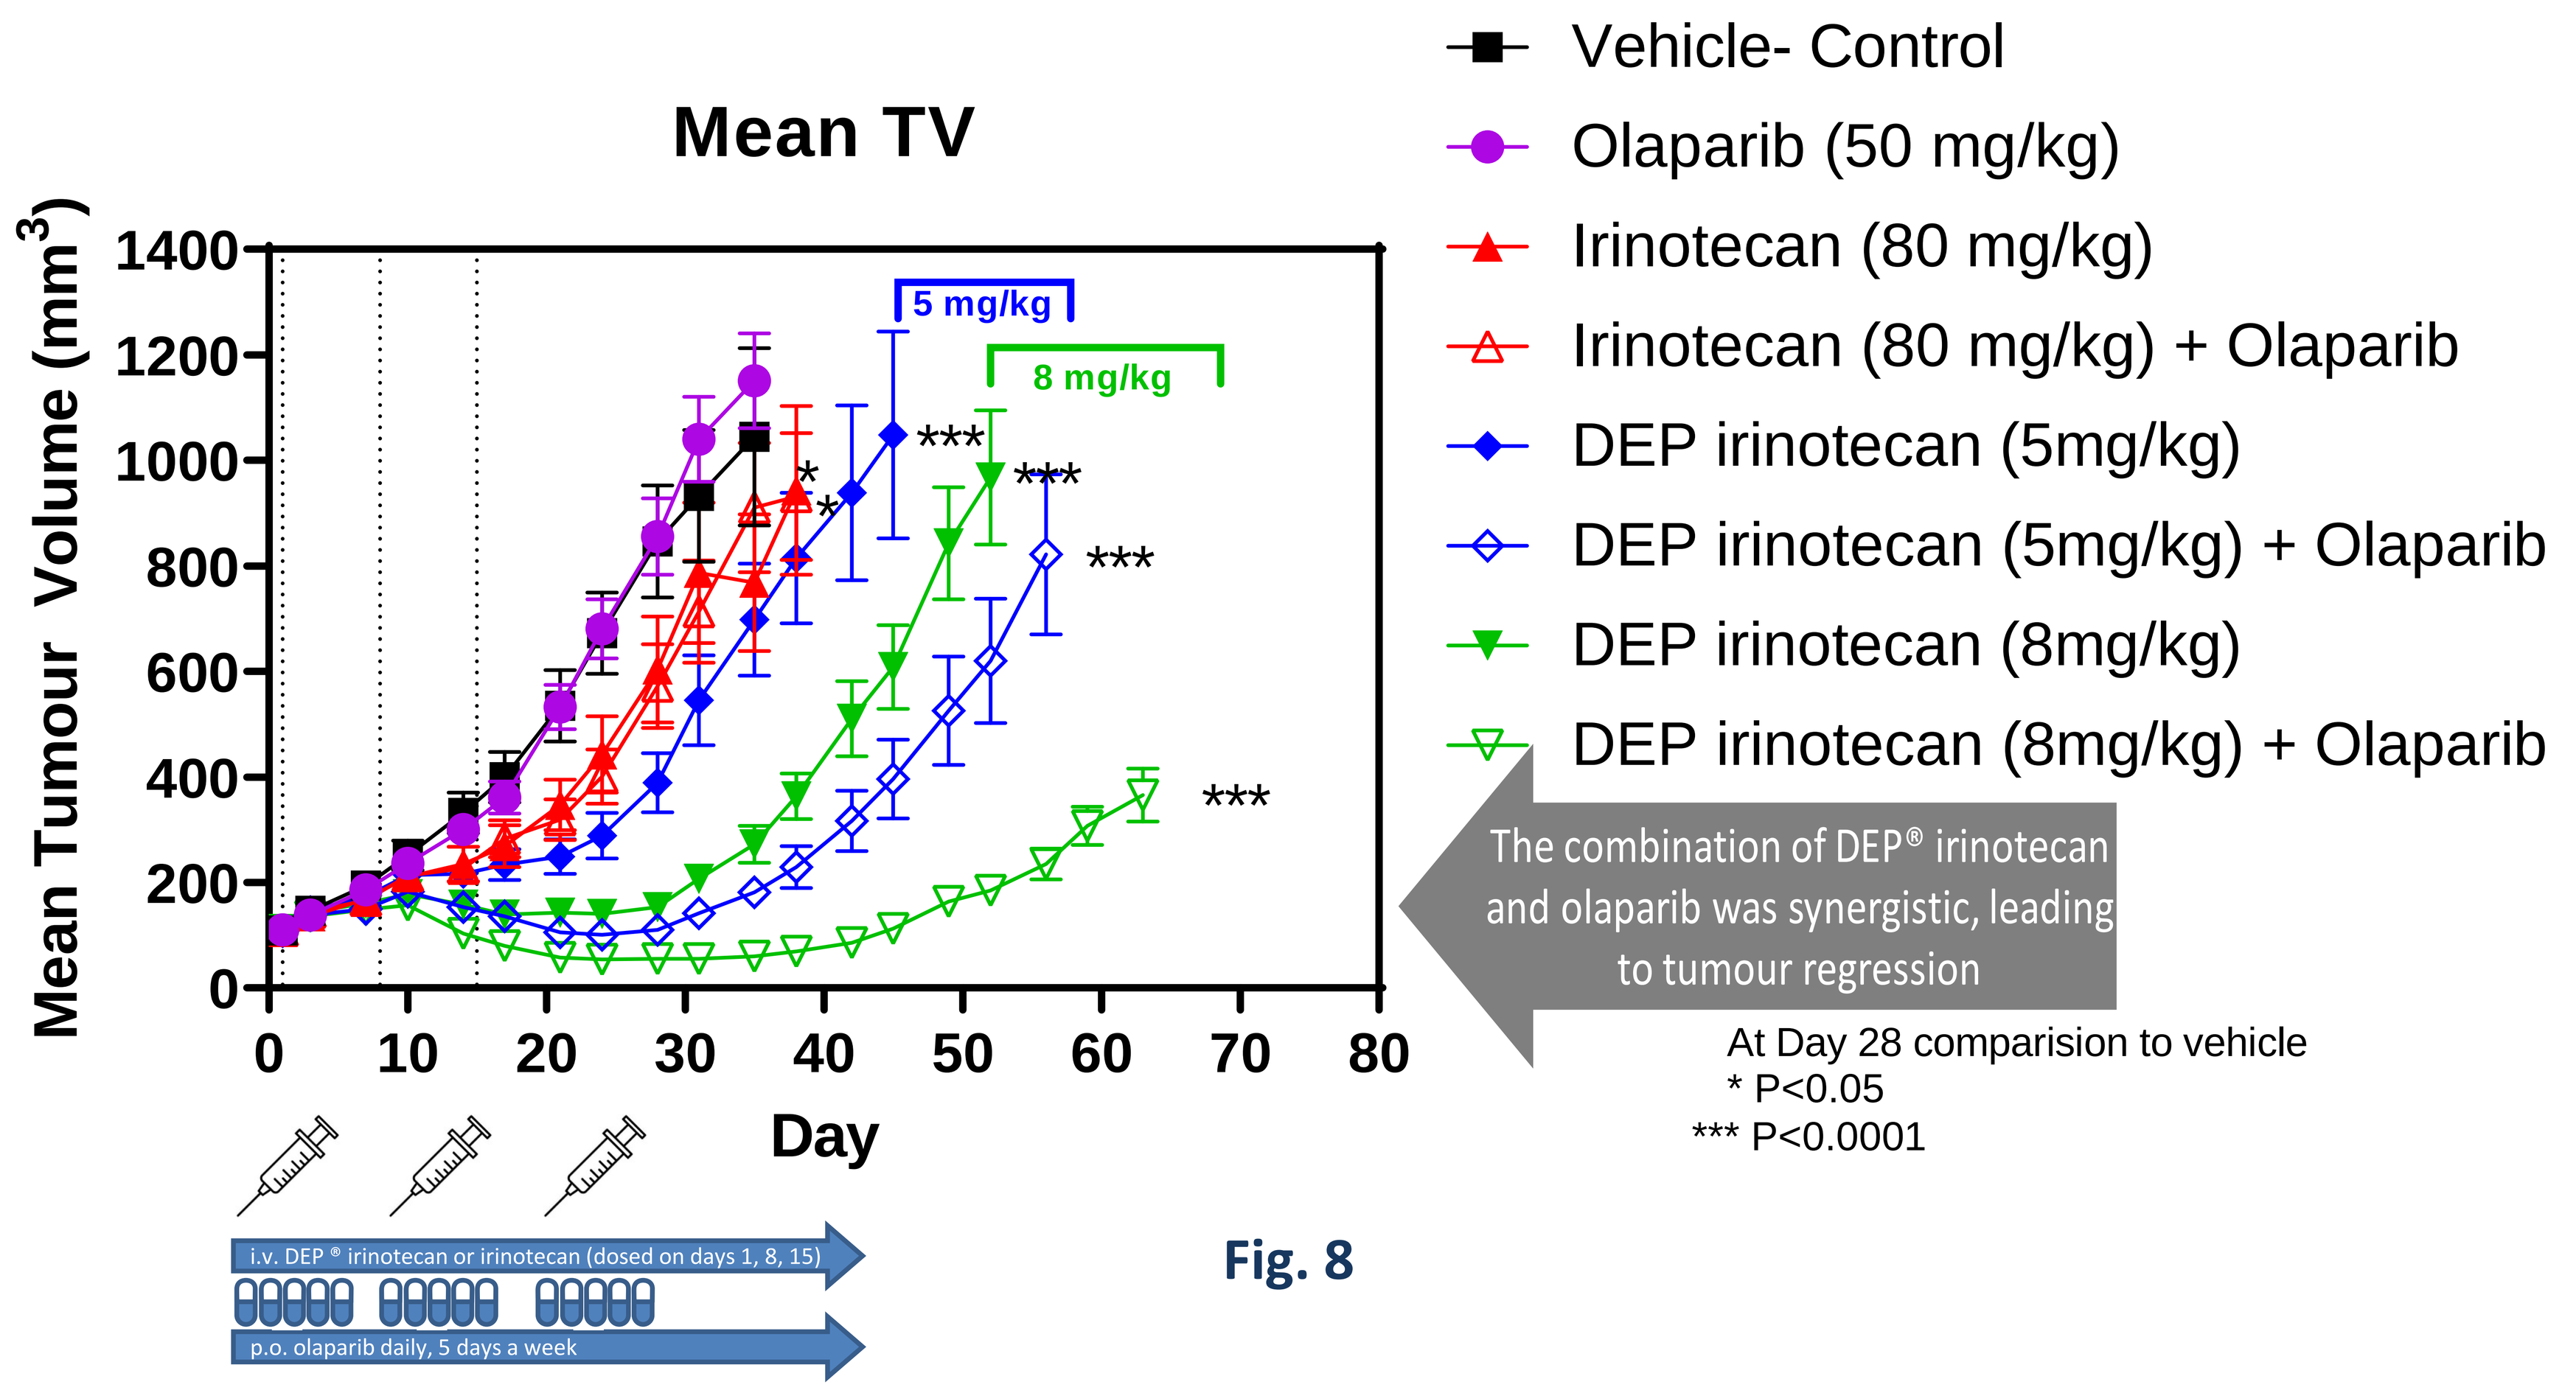
Mean TV
Mean Tumour Volume (mm³)
Day
5 mg/kg
8 mg/kg
Vehicle- Control
Olaparib (50 mg/kg)
Irinotecan (80 mg/kg)
Irinotecan (80 mg/kg) + Olaparib
DEP irinotecan (5mg/kg)
DEP irinotecan (5mg/kg) + Olaparib
DEP irinotecan (8mg/kg)
DEP irinotecan (8mg/kg) + Olaparib
The combination of DEP® irinotecan and olaparib was synergistic, leading to tumour regression
At Day 28 comparision to vehicle
* P<0.05
*** P<0.0001
i.v. DEP ® irinotecan or irinotecan (dosed on days 1, 8, 15)
p.o. olaparib daily, 5 days a week

Fig. 8

Olaparib, dosed either as a monotherapy, or dosed in combination with irinotecan, provided little effect on tumour growth. However, when olaparib was used in combination with DEP irinotecan, the enhanced anti-tumour activity observed was greater than the effect of each individual treatment alone. This statistically significant synergistic effect was observed in both the low-dose and high-dose DEP irinotecan + olaparib treated groups (P<0.0001).

Notwithstanding the statistical significance of the efficacy of DEP irinotecan alone, the addition of olaparib to DEP irinotecan provided an even greater level of tumour efficacy, with regression of tumours seen with both doses, as illustrated in figure 8 (P<0.0001). This was despite being dosed at significantly lower levels than the maximum tolerated dose (most efficacious dose). No other groups exhibited tumour regression in the refractory colon cancer model.

CLINICAL PROGRESS

DEP irinotecan is Starpharma's third internal DEP clinical candidate and recently completed the phase 1 portion of its development having met its objective of evaluating safety, tolerability, pharmacokinetics and preliminary efficacy data, and identifying a recommended phase 2 dose.

Results from the phase 1 study demonstrated that DEP irinotecan was well-tolerated and patients generally experienced less severe side effects, including no cases of severe diarrhea, which is particularly problematic (FDA black box warning) with the marketed form of irinotecan (Camptosar®)

Encouraging efficacy signals observed in 50% of evaluable patients to date – not only in patients with colorectal cancer, for which conventional irinotecan is approved, but also in patients with breast and pancreatic cancer. DEP irinotecan phase 2 clinical trial is currently underway at leading UK hospitals.